Controlling intracellular cortisol: Can HSD-1 inhibition reduce Cushing's syndrome morbidity and minimize adrenal insufficiency risk?

EP857

Frank S. Czerwiec[1], David A. Katz[1], and Paul M. Stewart [2]
[1]Sparrow Pharmaceuticals, Portland OR; [2]Faculty of Medicine & Health, University of Leeds, Leeds UK
contact: FCzerwiec@sparrowpharma.com

Endocrinologists focus on circulating and excreted cortisol for diagnosis of, and to assess severity and treatment response in, Cushing's syndrome (Cs). However, in Cs, morbidity is mediated by excess cortisol binding to intracellular glucocorticoid (GC), mineralocorticoid (MC), and non-genomic receptors.

We and others have demonstrated that 11β-hydroxysteroid dehydrogenase type 1 (HSD-1) is the source of about half of intrahepatocellular cortisol in healthy adults, patients with diabetes, and patients with mild hypercortisolism. The circulating concentration of cortisol is higher than cortisone, yet its bioavailability is limited, due to the former's greater affinity to plasma proteins. Free circulating cortisol and cortisone concentrations are similar, the latter providing a reservoir of "inactive" GC that enters cells and is rapidly converted by HSD-1 to cortisol. Patients with severe hypercortisolism who are deficient in HSD-1 activity showed no cortisol-related morbidity. A HSD-1 inhibitor prevented the deleterious effects of prednisolone on glycemic control and osteocalcin in a Phase 1 clinical trial. Mice without the *Hsd11b1* gene (which encodes HSD-1) or treated with a HSD-1 inhibitor were protected from glucose intolerance, hyperinsulinemia, hepatic steatosis, adiposity, hypertension, myopathy, dermal atrophy, and trabecular bone loss associated with GC administration.

In states of glucocorticoid excess, liver HSD-1 activity is enhanced, as assessed by the HSD-1 ratio of urinary excreted cortisol/cortisone metabolites. Administration of a HSD-1 inhibitor to such patients could reduce Cs morbidity via reduction of the cortisol (or HSD-1 metabolized medications, e.g., prednisolone) available to intracellular receptors. Furthermore, unlike current Cs treatments that are associated with substantial adrenal insufficiency risk, under full and sustained HSD-1 inhibition autonomously produced or ACTH-stimulated cortisol remains elevated in the circulatory pool, available to enter cells and act at GC, MC or non-genomic receptors at concentrations likely sufficient to prevent adrenal insufficiency. Those model predictions are under evaluation in ongoing Phase 2 clinical trials of the HSD-1 inhibitor SPI-62 in patients with ACTH-dependent Cushing's syndrome, autonomous cortisol secretion, and (in combination with prednisolone) polymyalgia rheumatica.

Until further direct experience in these conditions is gathered, we still advise caution in patients with modest autonomous cortisol secretion, who may have suppressed ACTH and atrophied normal adrenal tissue, particularly in situations where stress-dose, non-precursor GC steroids should be considered (e.g., use hydrocortisone not cortisone acetate, or prednisolone not prednisone).

Adrenal vein cortisol and cortisone effluent (8:1 ratio)

HSD-1 is primarily in liver, but also found in adipose, brain, skin, bone, muscle, blood vessels

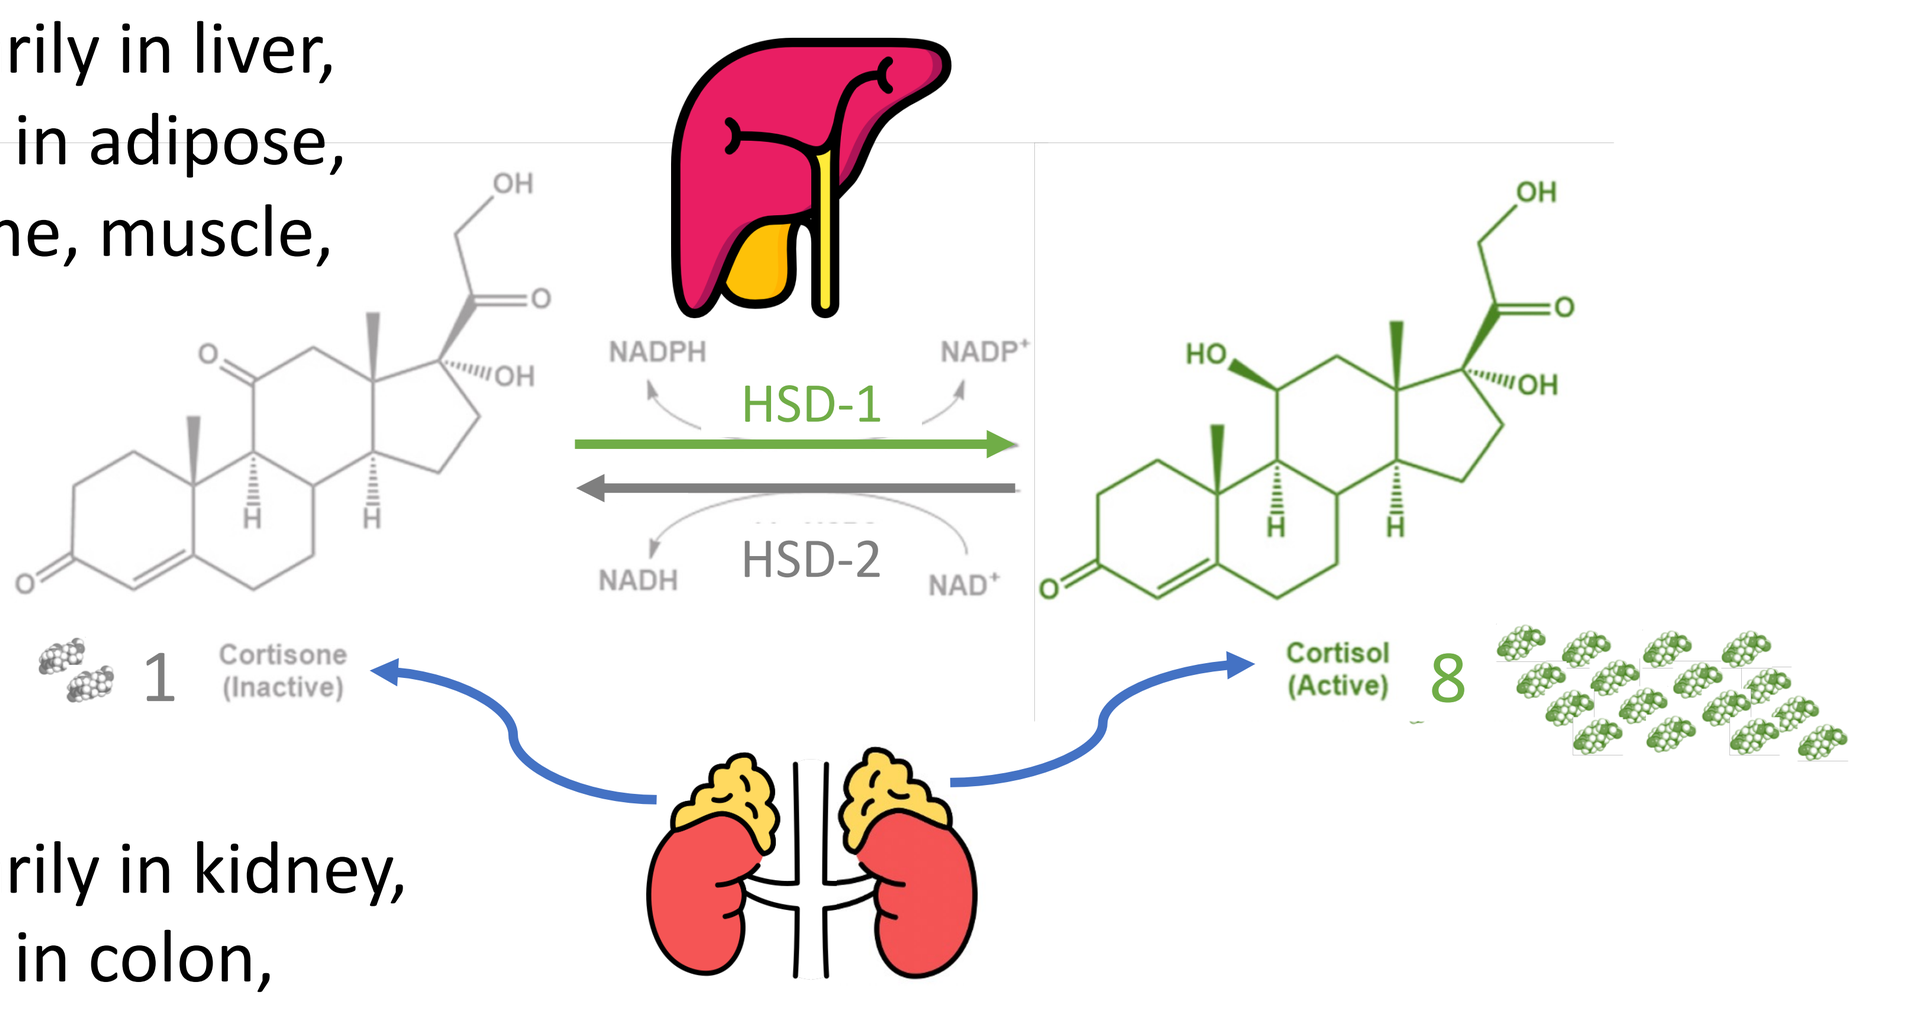
NADPH
NADP+
HSD-1
HSD-2
NADH
NAD+
1 Cortisone (Inactive)
Cortisol (Active) 8

HSD-2 is primarily in kidney, but also found in colon, lung, salivary gland

Total glucocorticoid production 17.3 mmol/24hr
(est. by excretion of cortisol, cortisone and their metabolites)

With SPI-62, GC production increases to 47.9 mmol/24hr (277%) and ACTH increases of 56, 69, 29, and 91% at 2, 4, 12, 24 hr. post-dose. Adrenal androgens and end products (DHEA, DHEA-S, and in women testosterone, but not estrogen) increased but within the normal range)

Hepatic HSD-1 and renal HSD-2 drive plasma cortisol:cortisone equilibrium

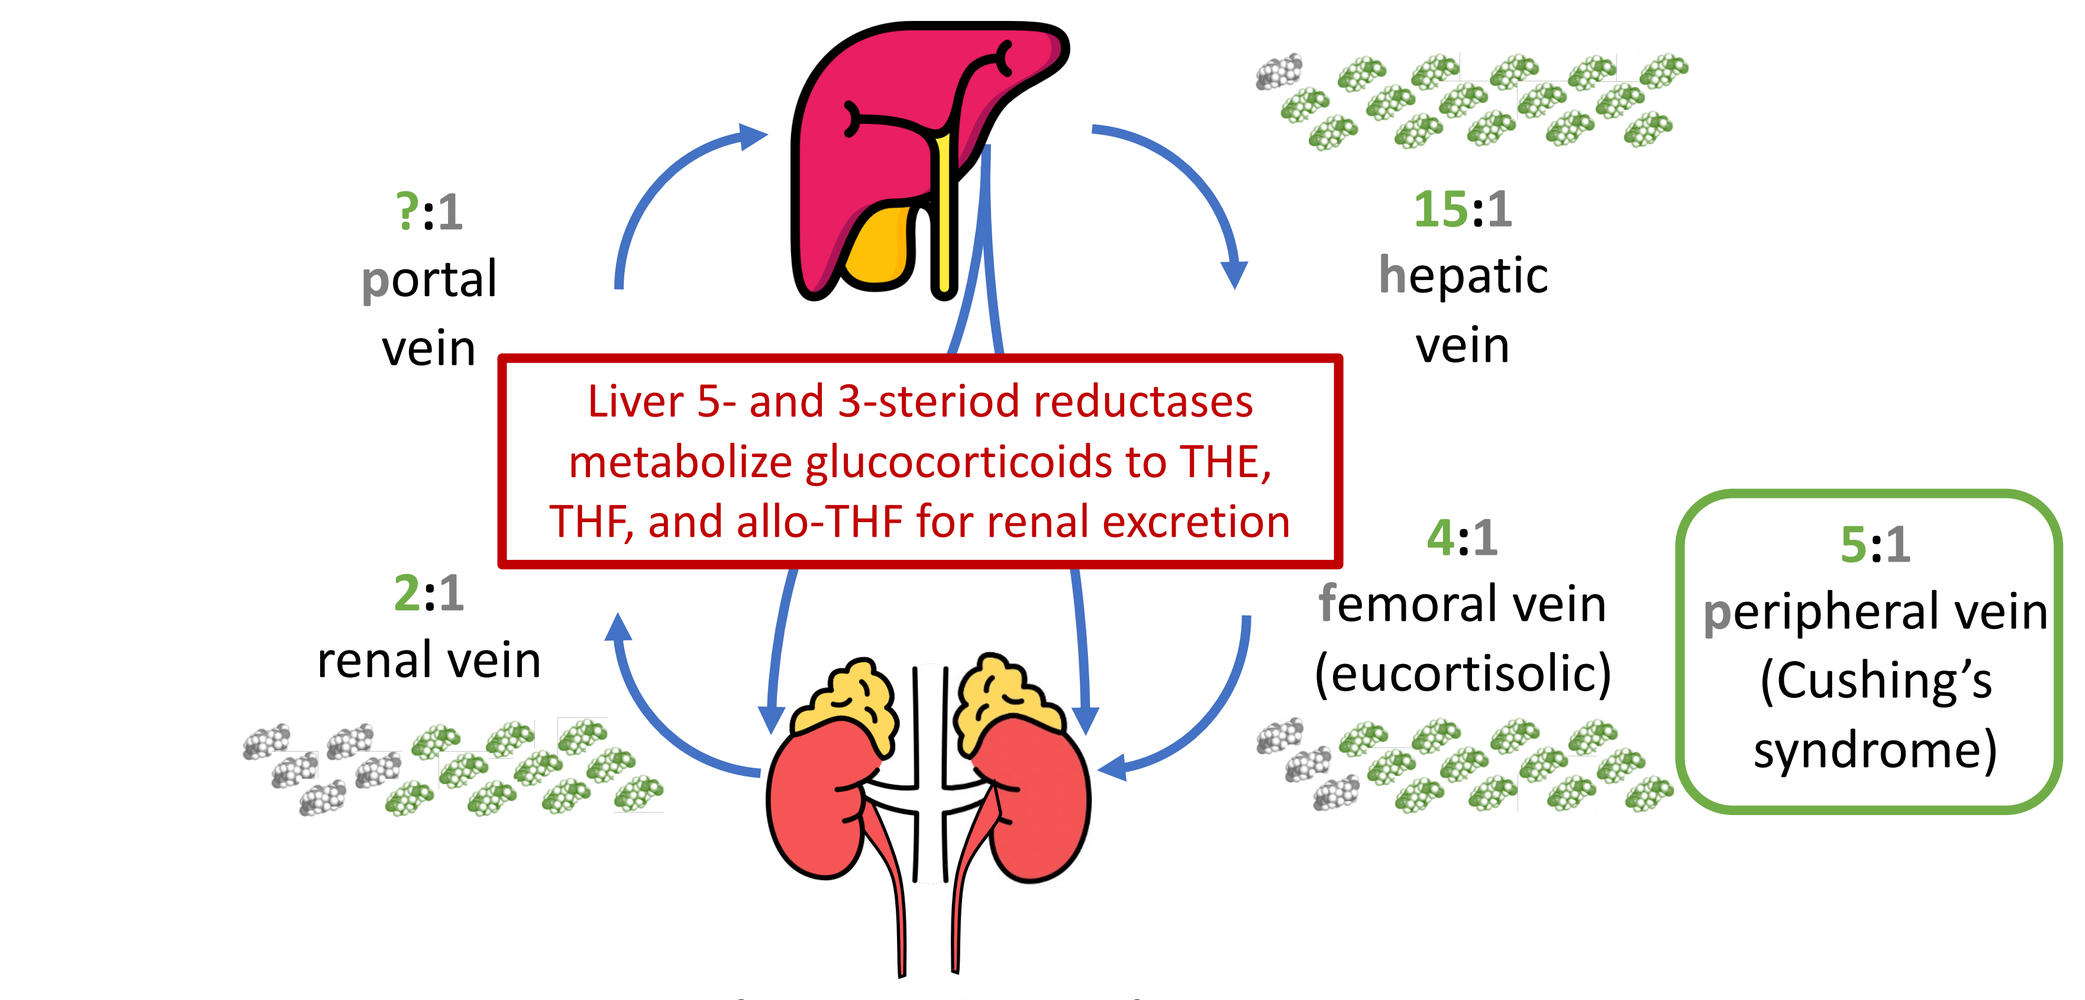
?:1
portal vein
15:1
hepatic vein
Liver 5- and 3-steriod reductases metabolize glucocorticoids to THE, THF, and allo-THF for renal excretion
2:1
renal vein
4:1
femoral vein (eucortisolic)
5:1
peripheral vein (Cushing's syndrome)

Tetra-hydro cortisol = THF
Tetra-hydro cortisone = THE

Urinary HSD-1 ratio:
(THF + allo-THF) / THE ≅ 1

HSD-1 Ratio (Normal 1:1)

Control HSD-1 Ratio = (4.4 + 4.1)/8.5 mmol/L ≅ 1.0 to 1.1
SPI-62 HSD-1 Ratio = (2.1 = 2.8)/43 mmol/L ≅ 0.6 to 0.09

HSD-1 Ratio (suppressed 1:10)

NOTE: Even with an ACTH-induced total glucocorticoid rise from 17.3 to 47.9 mmol/24hr (277%), SPI-62 reduced cortisol metabolites by 40-50% and increased cortisone metabolites 5-fold

Plasma protein binding and end-organ HSD-1 drive intracellular cortisol:cortisone ratio and receptor activation

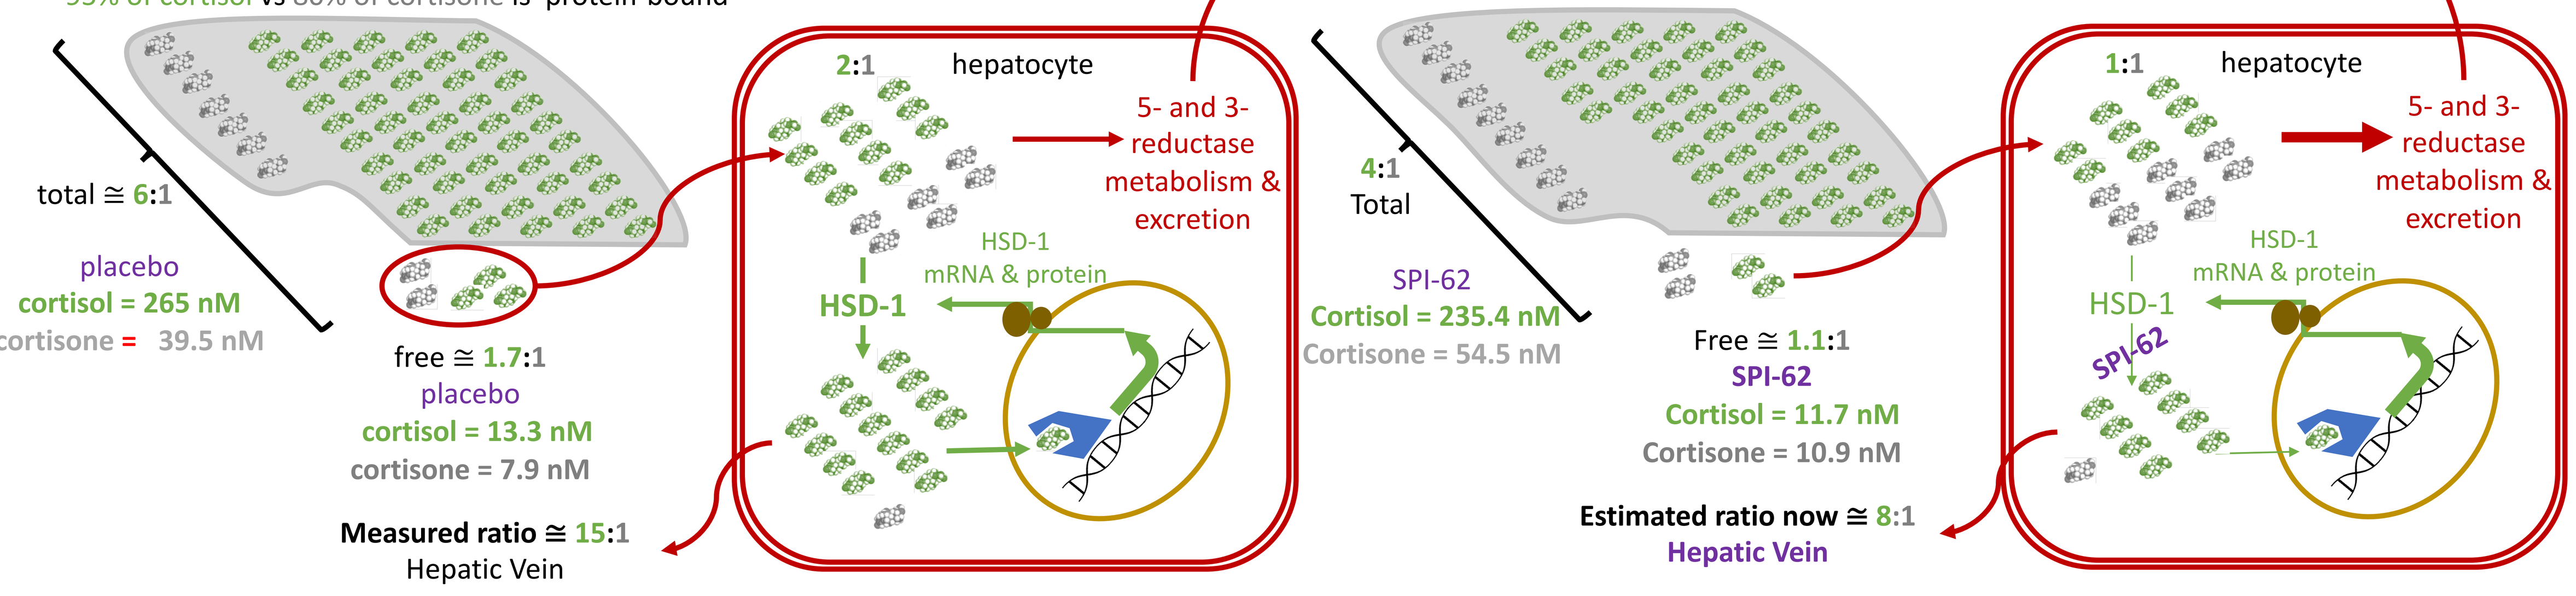
95% of cortisol vs 80% of cortisone is protein-bound
total ≅ 6:1
placebo
cortisol = 265 nM
cortisone = 39.5 nM
free ≅ 1.7:1
placebo
cortisol = 13.3 nM
cortisone = 7.9 nM
Measured ratio ≅ 15:1
Hepatic Vein
2:1
hepatocyte
5- and 3- reductase metabolism & excretion
HSD-1 mRNA & protein
HSD-1
4:1
Total
SPI-62
Cortisol = 235.4 nM
Cortisone = 54.5 nM
Free ≅ 1.1:1
SPI-62
Cortisol = 11.7 nM
Cortisone = 10.9 nM
Estimated ratio now ≅ 8:1
Hepatic Vein
1:1
hepatocyte
5- and 3- reductase metabolism & excretion
HSD-1 mRNA & protein
HSD-1
SPI-62

PURPLE represents Sparrow Pharmaceuticals' data on file

References: Xin Yang, et al., 2018 Drug Metab. Disposition; Stuart A Morgan, et al., 2014 PNAS; Morita H, et al., 2004 Metabolism; Jeremy Cohen, et al., 2012 Shock; Xin Yang, et al., 2018 Drug Metab. Disposition; Stuart A Morgan, et al., 2014 PNAS; Morita H, et al., 2004 Metabolism, Theiler-Schwetz V. et al., 2023 Curr Opin Endo Diab Obesity

Healthy

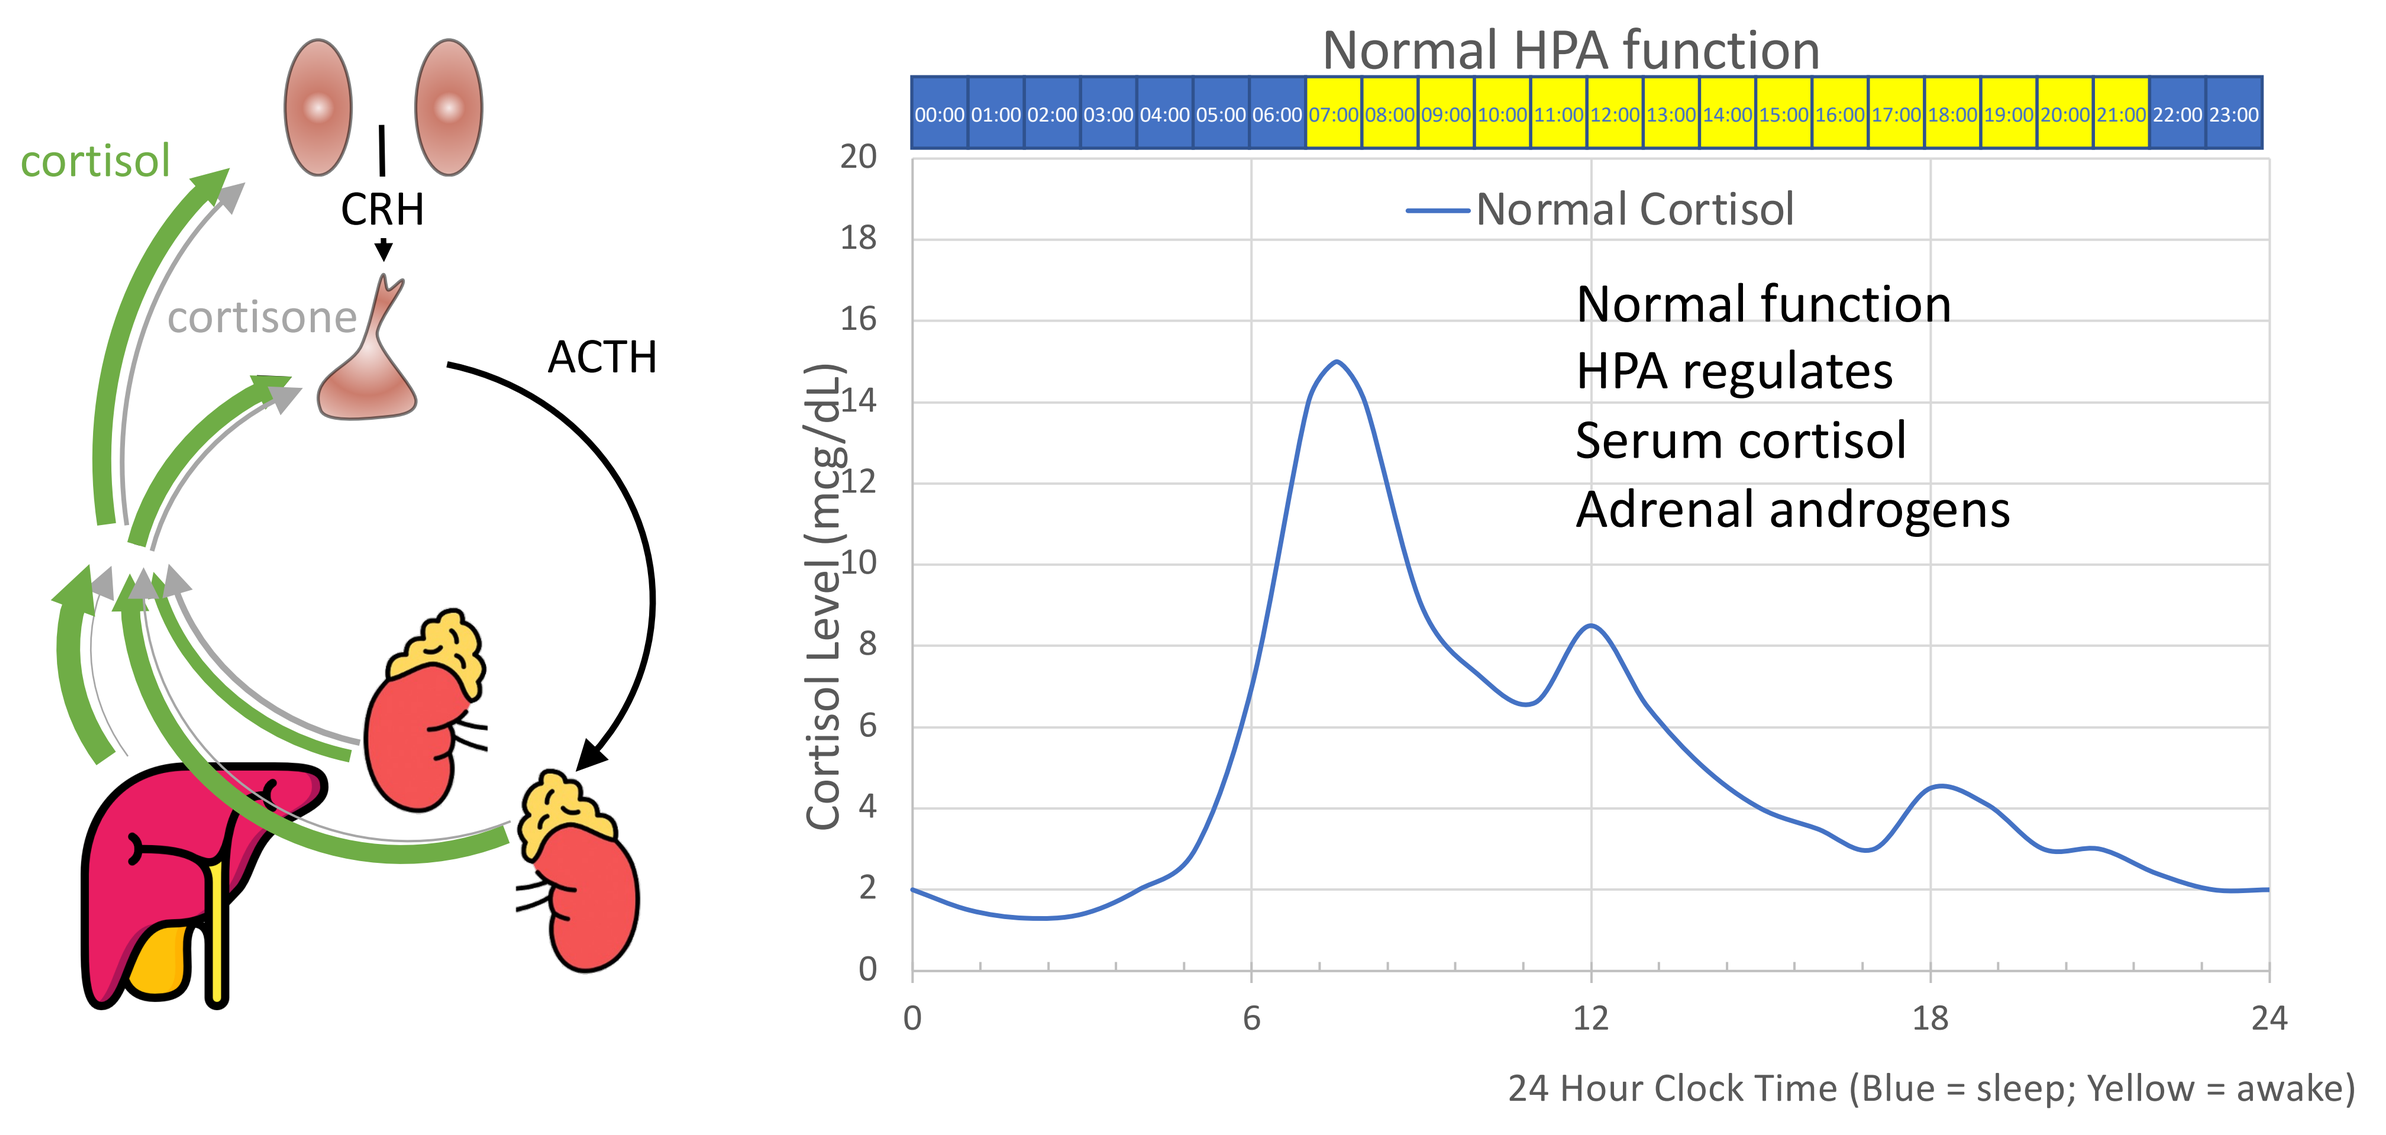
cortisol
CRH
cortisone
ACTH
Normal HPA function
Normal Cortisol
Normal function
HPA regulates
Serum cortisol
Adrenal androgens
Cortisol Level (mcg/dL)
24 Hour Clock Time (Blue = sleep; Yellow = awake)

Autonomous Cortisol Secretion (ACS)

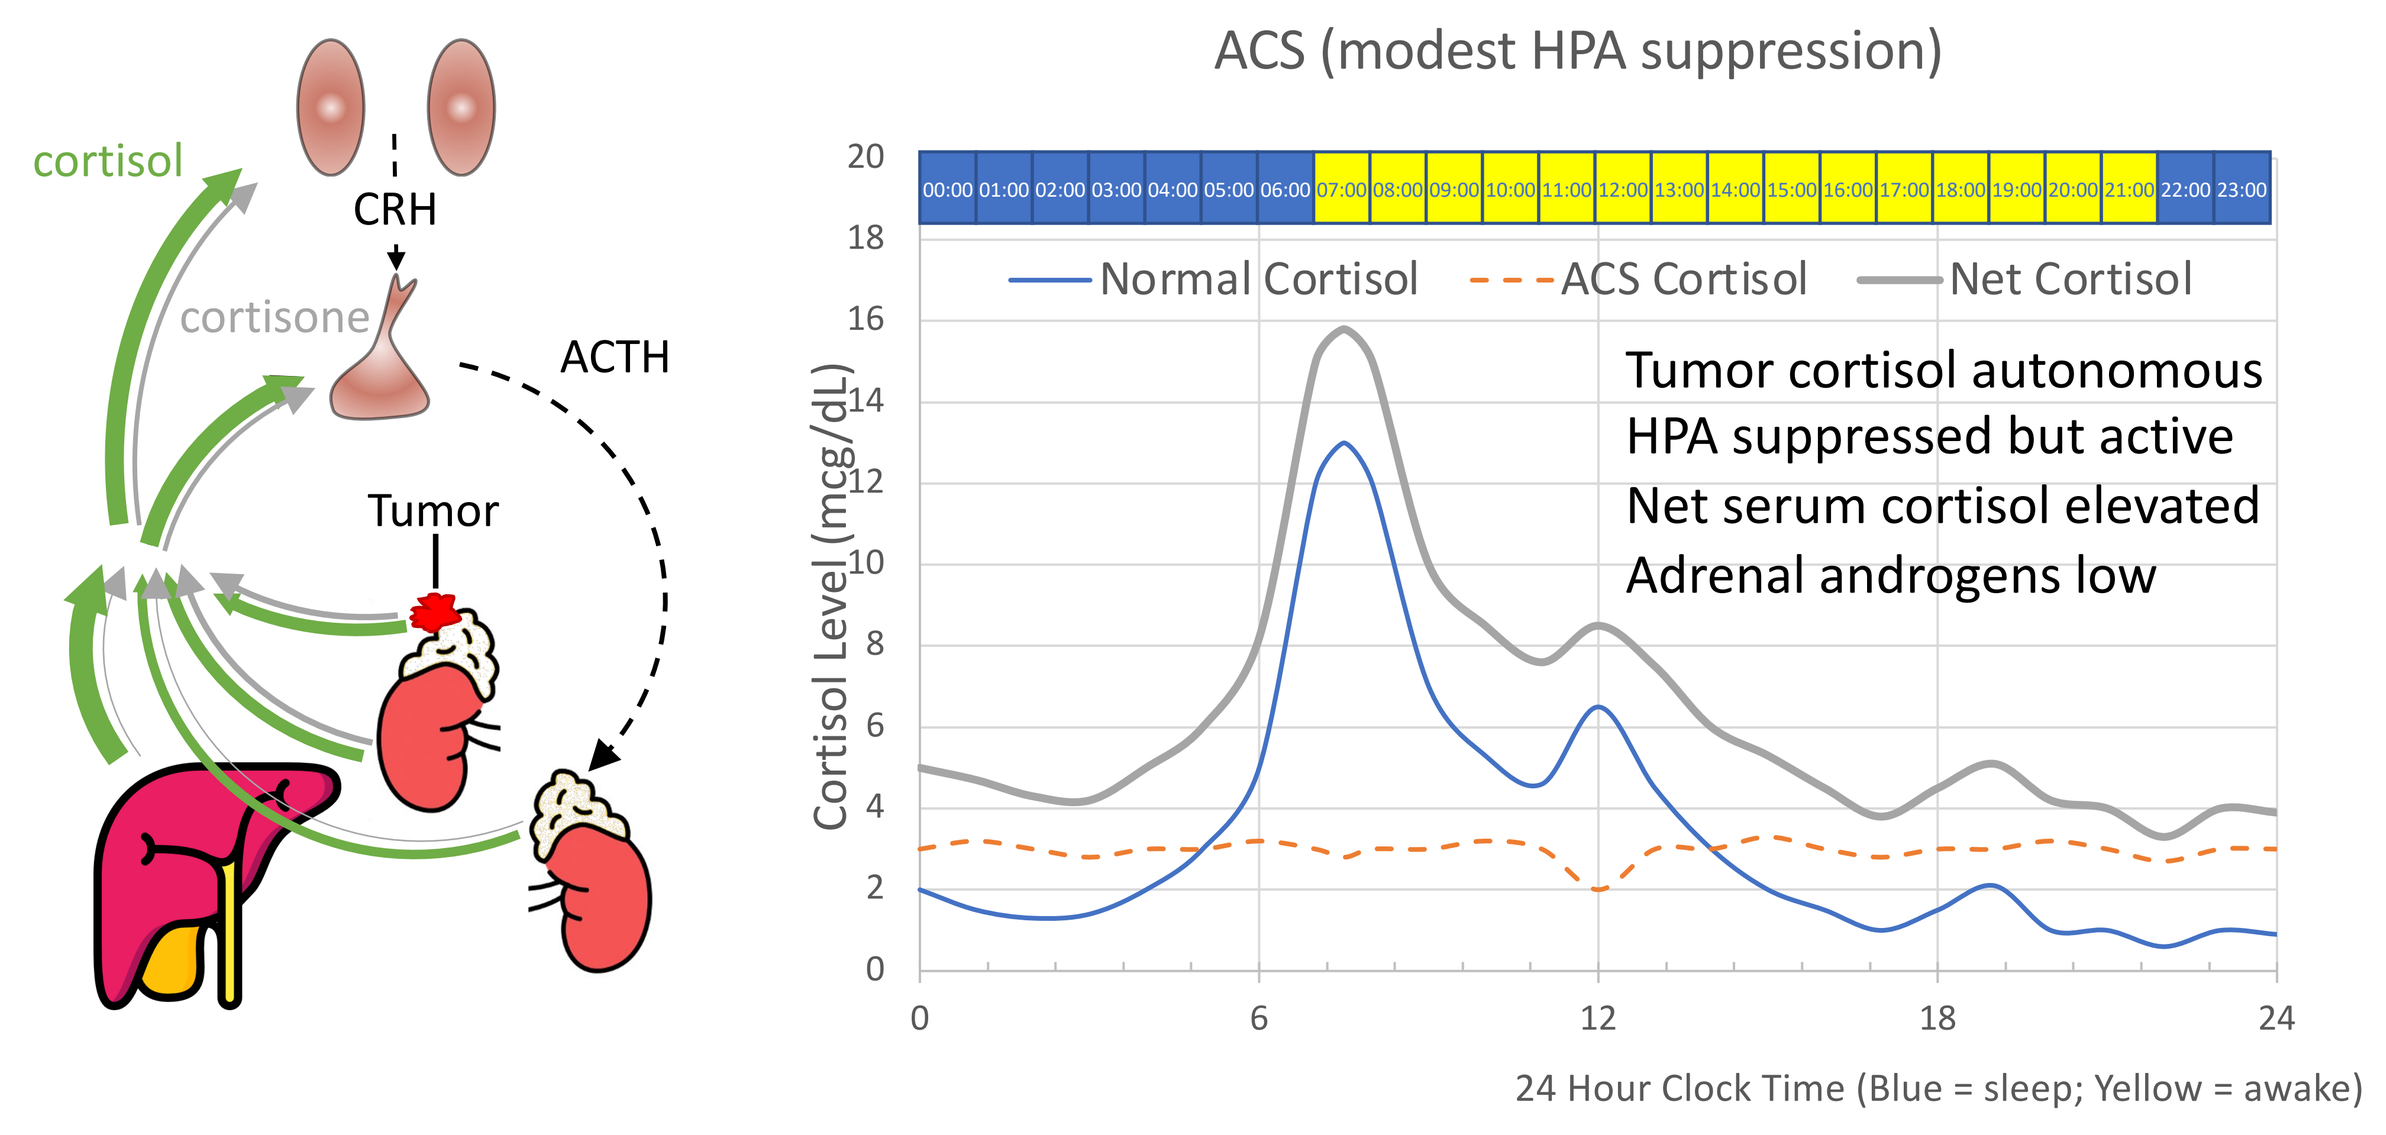
cortisol
CRH
cortisone
ACTH
Tumor
ACS (modest HPA suppression)
Normal Cortisol
ACS Cortisol
Net Cortisol
Tumor cortisol autonomous
HPA suppressed but active
Net serum cortisol elevated
Adrenal androgens low
Cortisol Level (mcg/dL)
24 Hour Clock Time (Blue = sleep; Yellow = awake)

ACTH-driven Cushing's syndrome

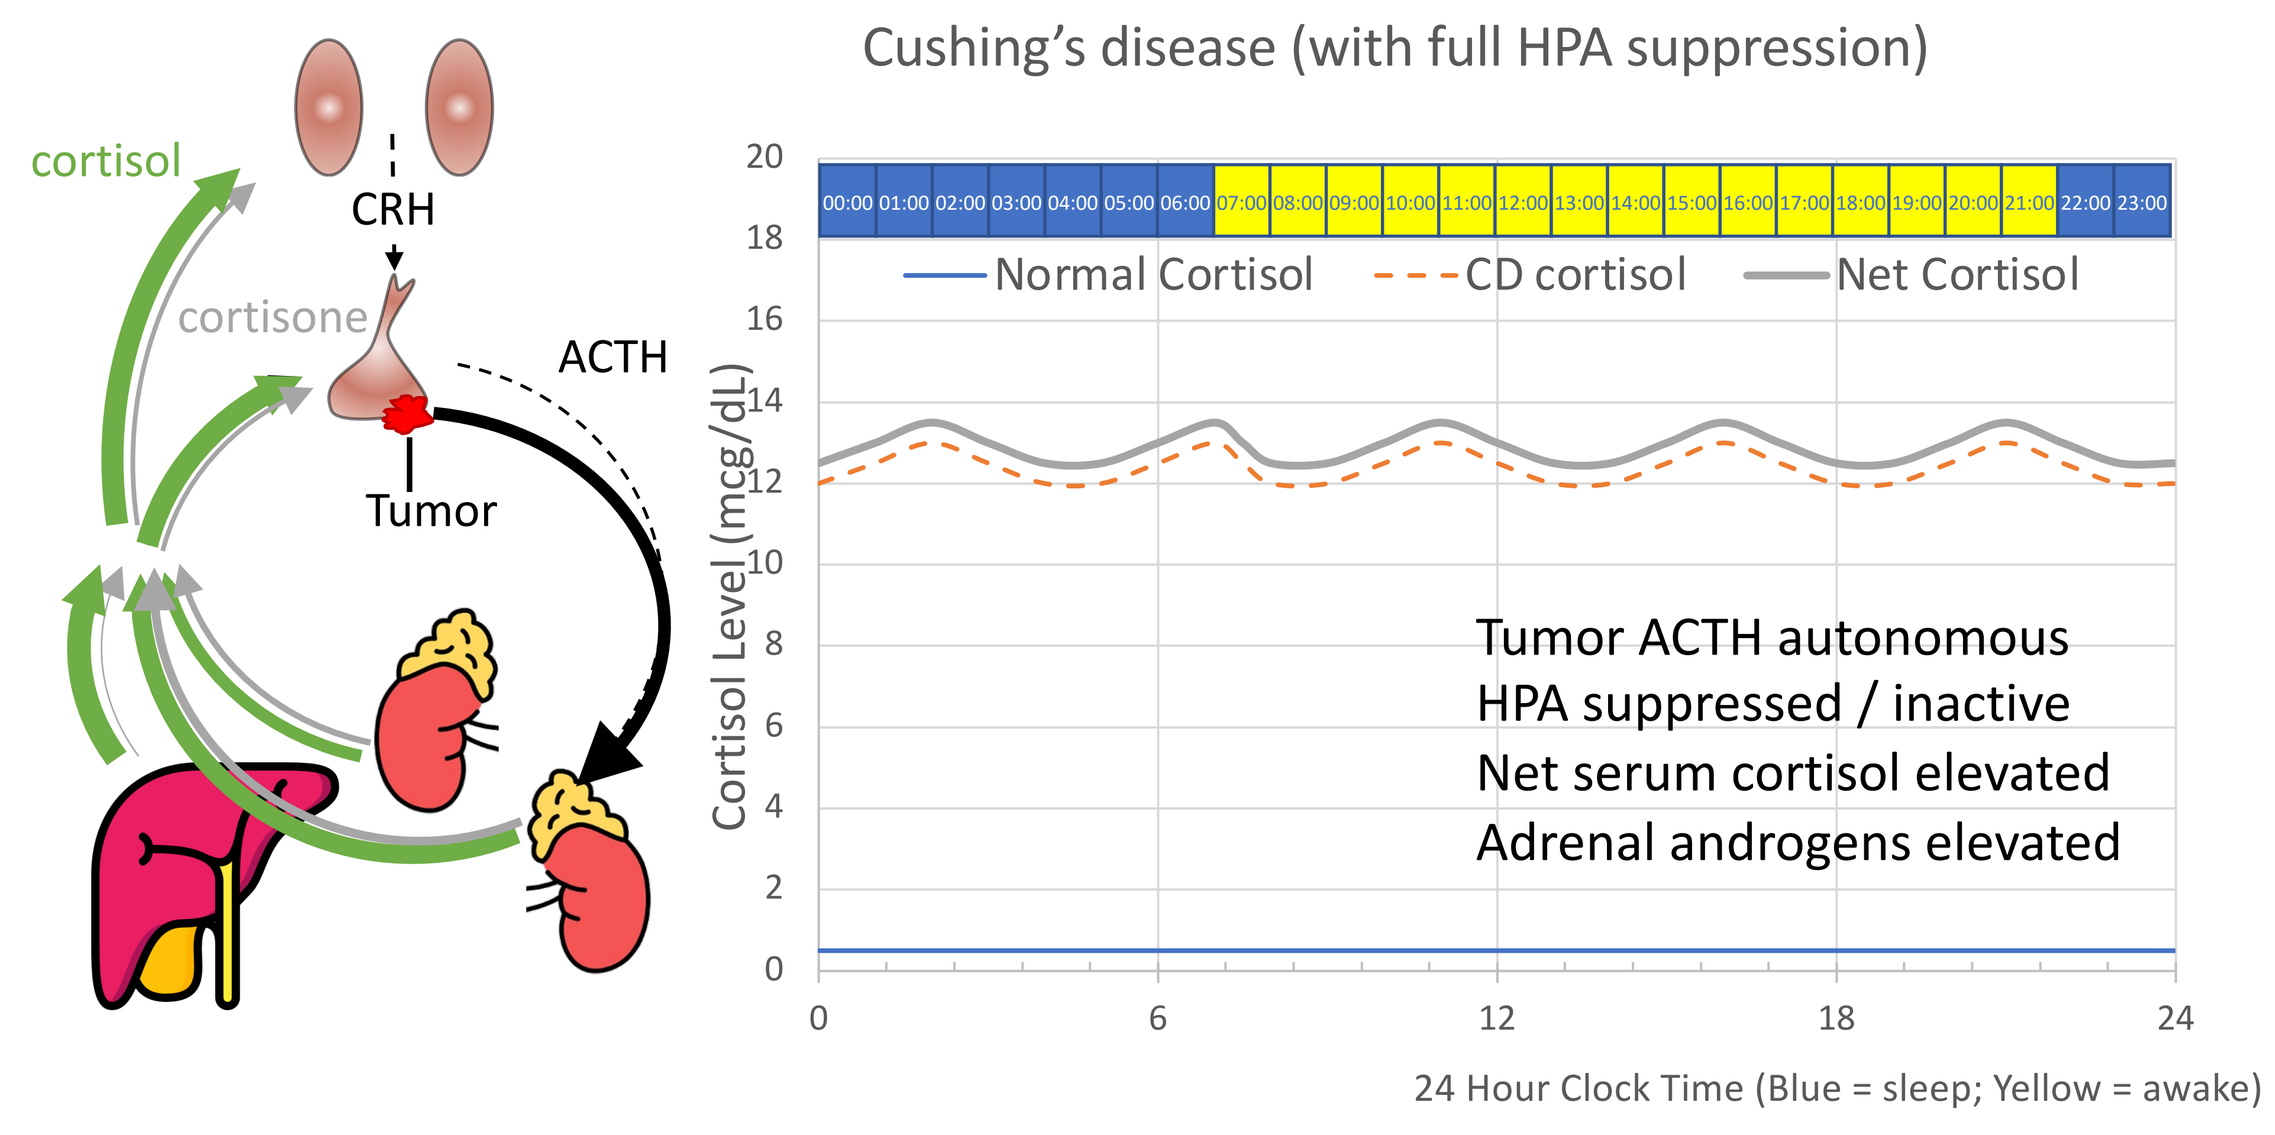
cortisol
CRH
cortisone
ACTH
Tumor
Cushing's disease (with full HPA suppression)
Normal Cortisol
CD cortisol
Net Cortisol
Tumor ACTH autonomous
HPA suppressed / inactive
Net serum cortisol elevated
Adrenal androgens elevated
Cortisol Level (mcg/dL)
24 Hour Clock Time (Blue = sleep; Yellow = awake)

Healthy + SPI-62

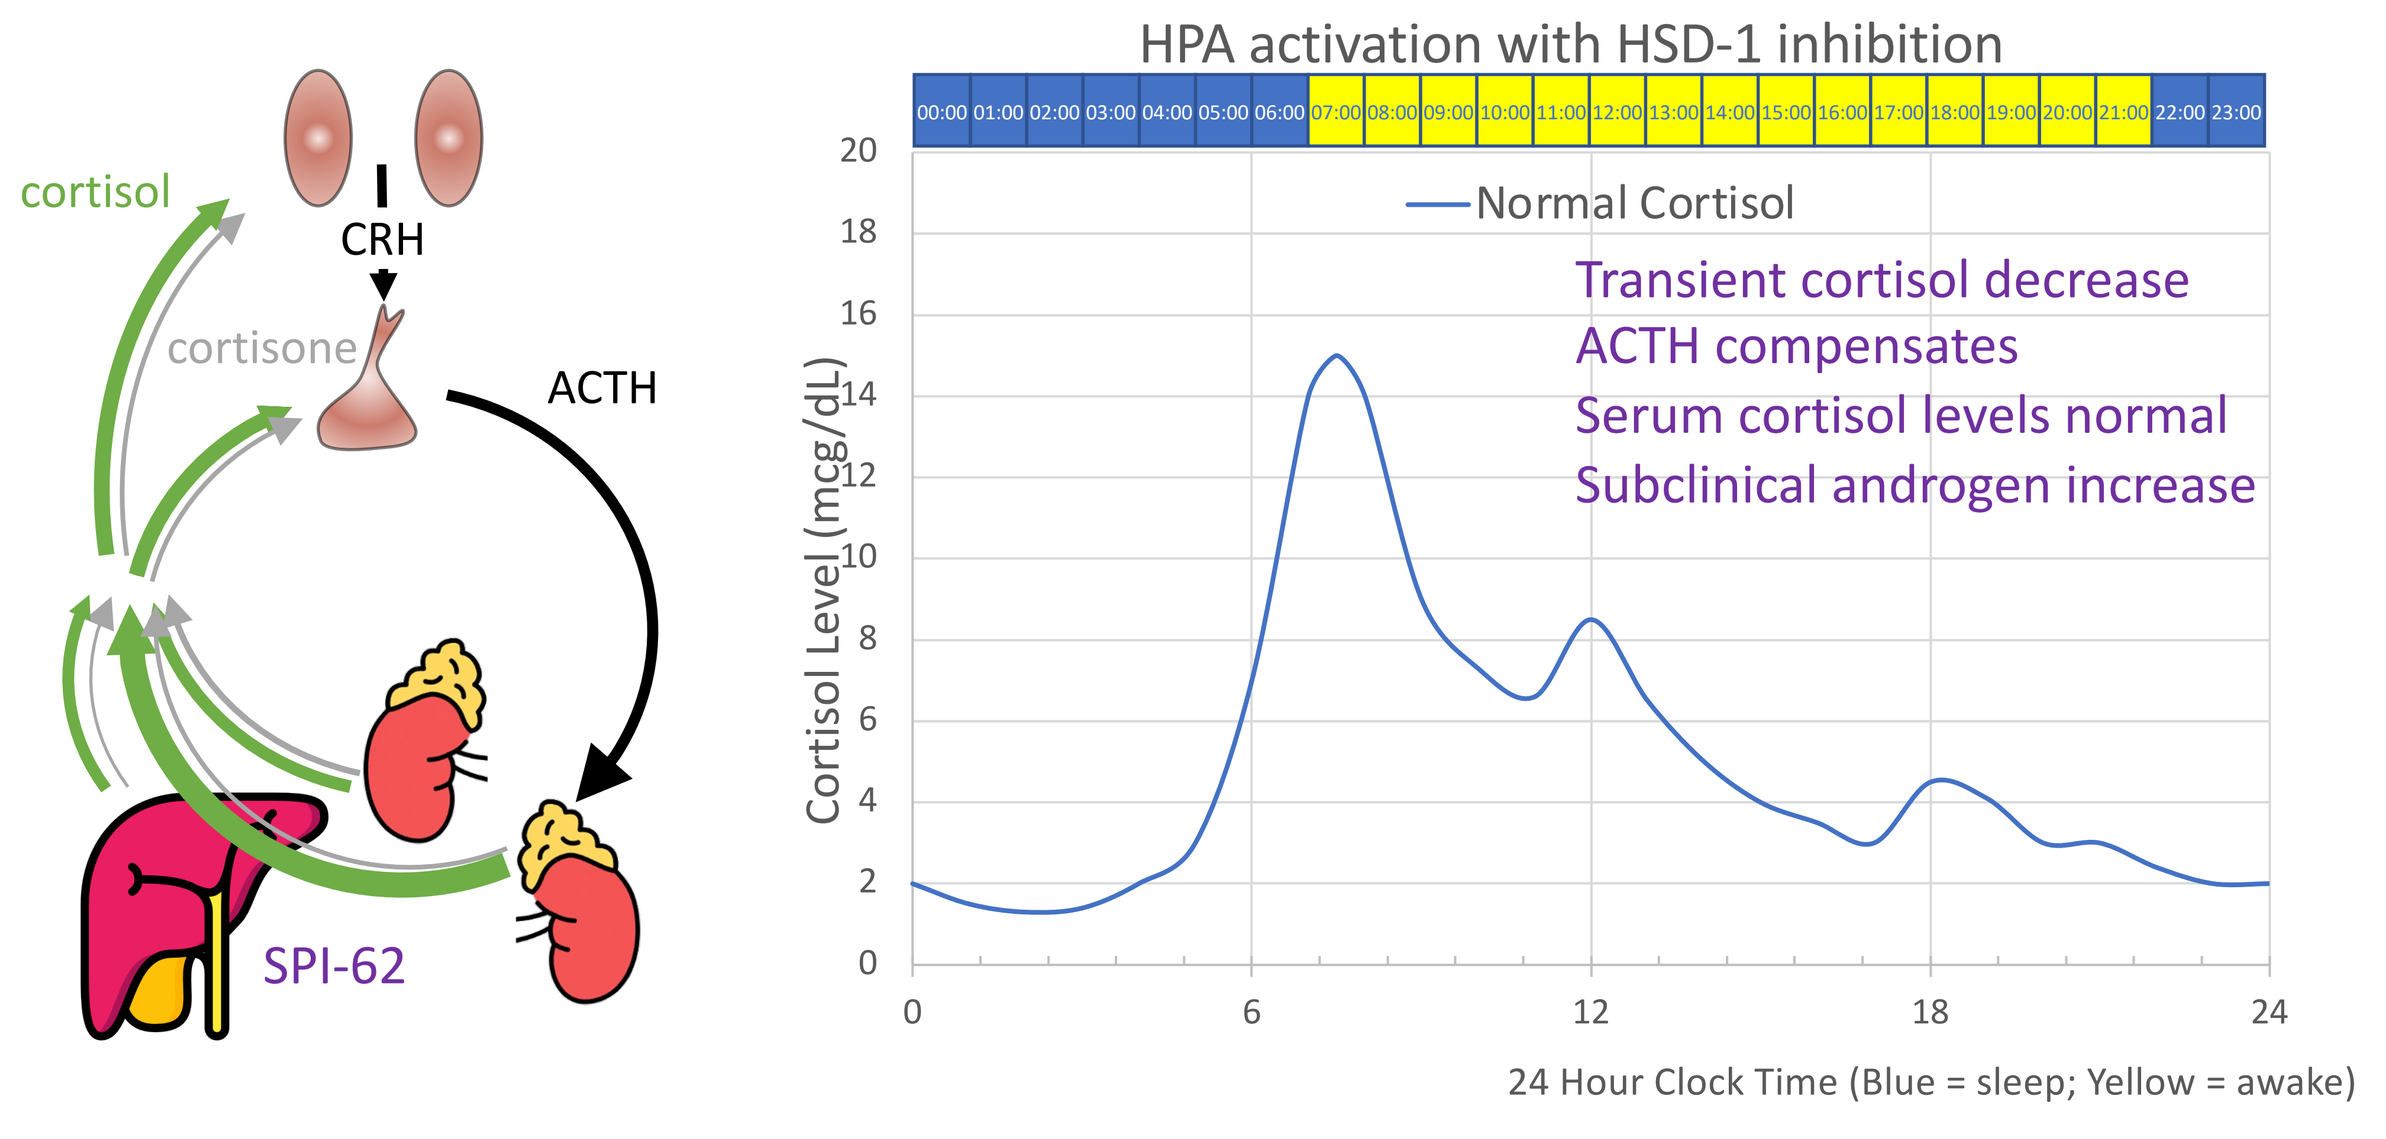
cortisol
CRH
cortisone
ACTH
SPI-62
HPA activation with HSD-1 inhibition
Normal Cortisol
Transient cortisol decrease
ACTH compensates
Serum cortisol levels normal
Subclinical androgen increase
Cortisol Level (mcg/dL)
24 Hour Clock Time (Blue = sleep; Yellow = awake)

ACS + SPI-62

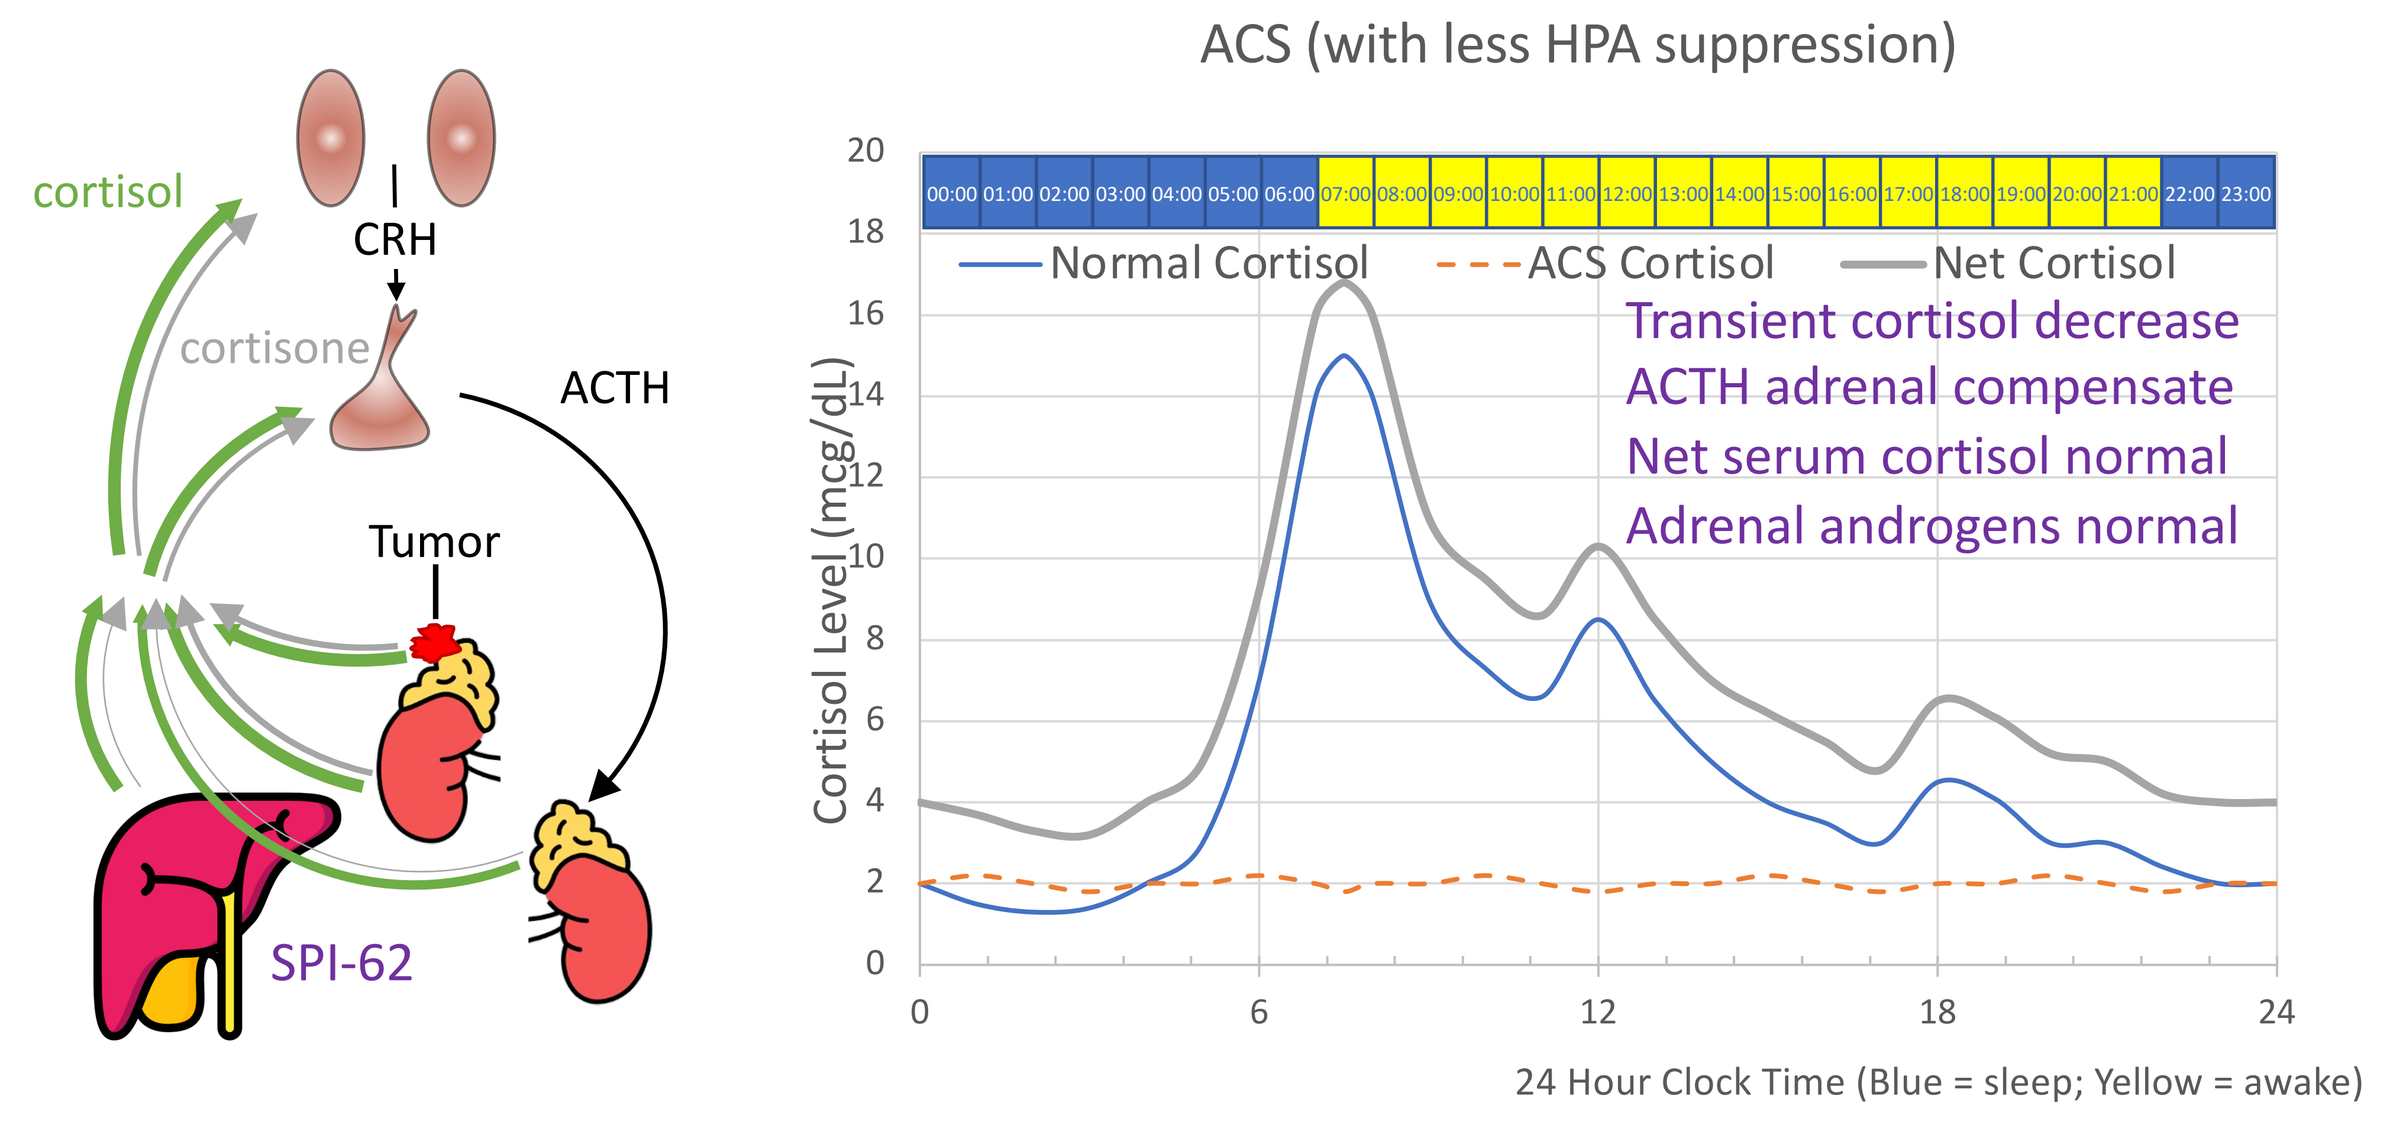
cortisol
CRH
cortisone
ACTH
Tumor
SPI-62
ACS (with less HPA suppression)
Normal Cortisol
ACS Cortisol
Net Cortisol
Transient cortisol decrease
ACTH adrenal compensate
Net serum cortisol normal
Adrenal androgens normal
Cortisol Level (mcg/dL)
24 Hour Clock Time (Blue = sleep; Yellow = awake)

ACTH-driven Cushing's + SPI-62

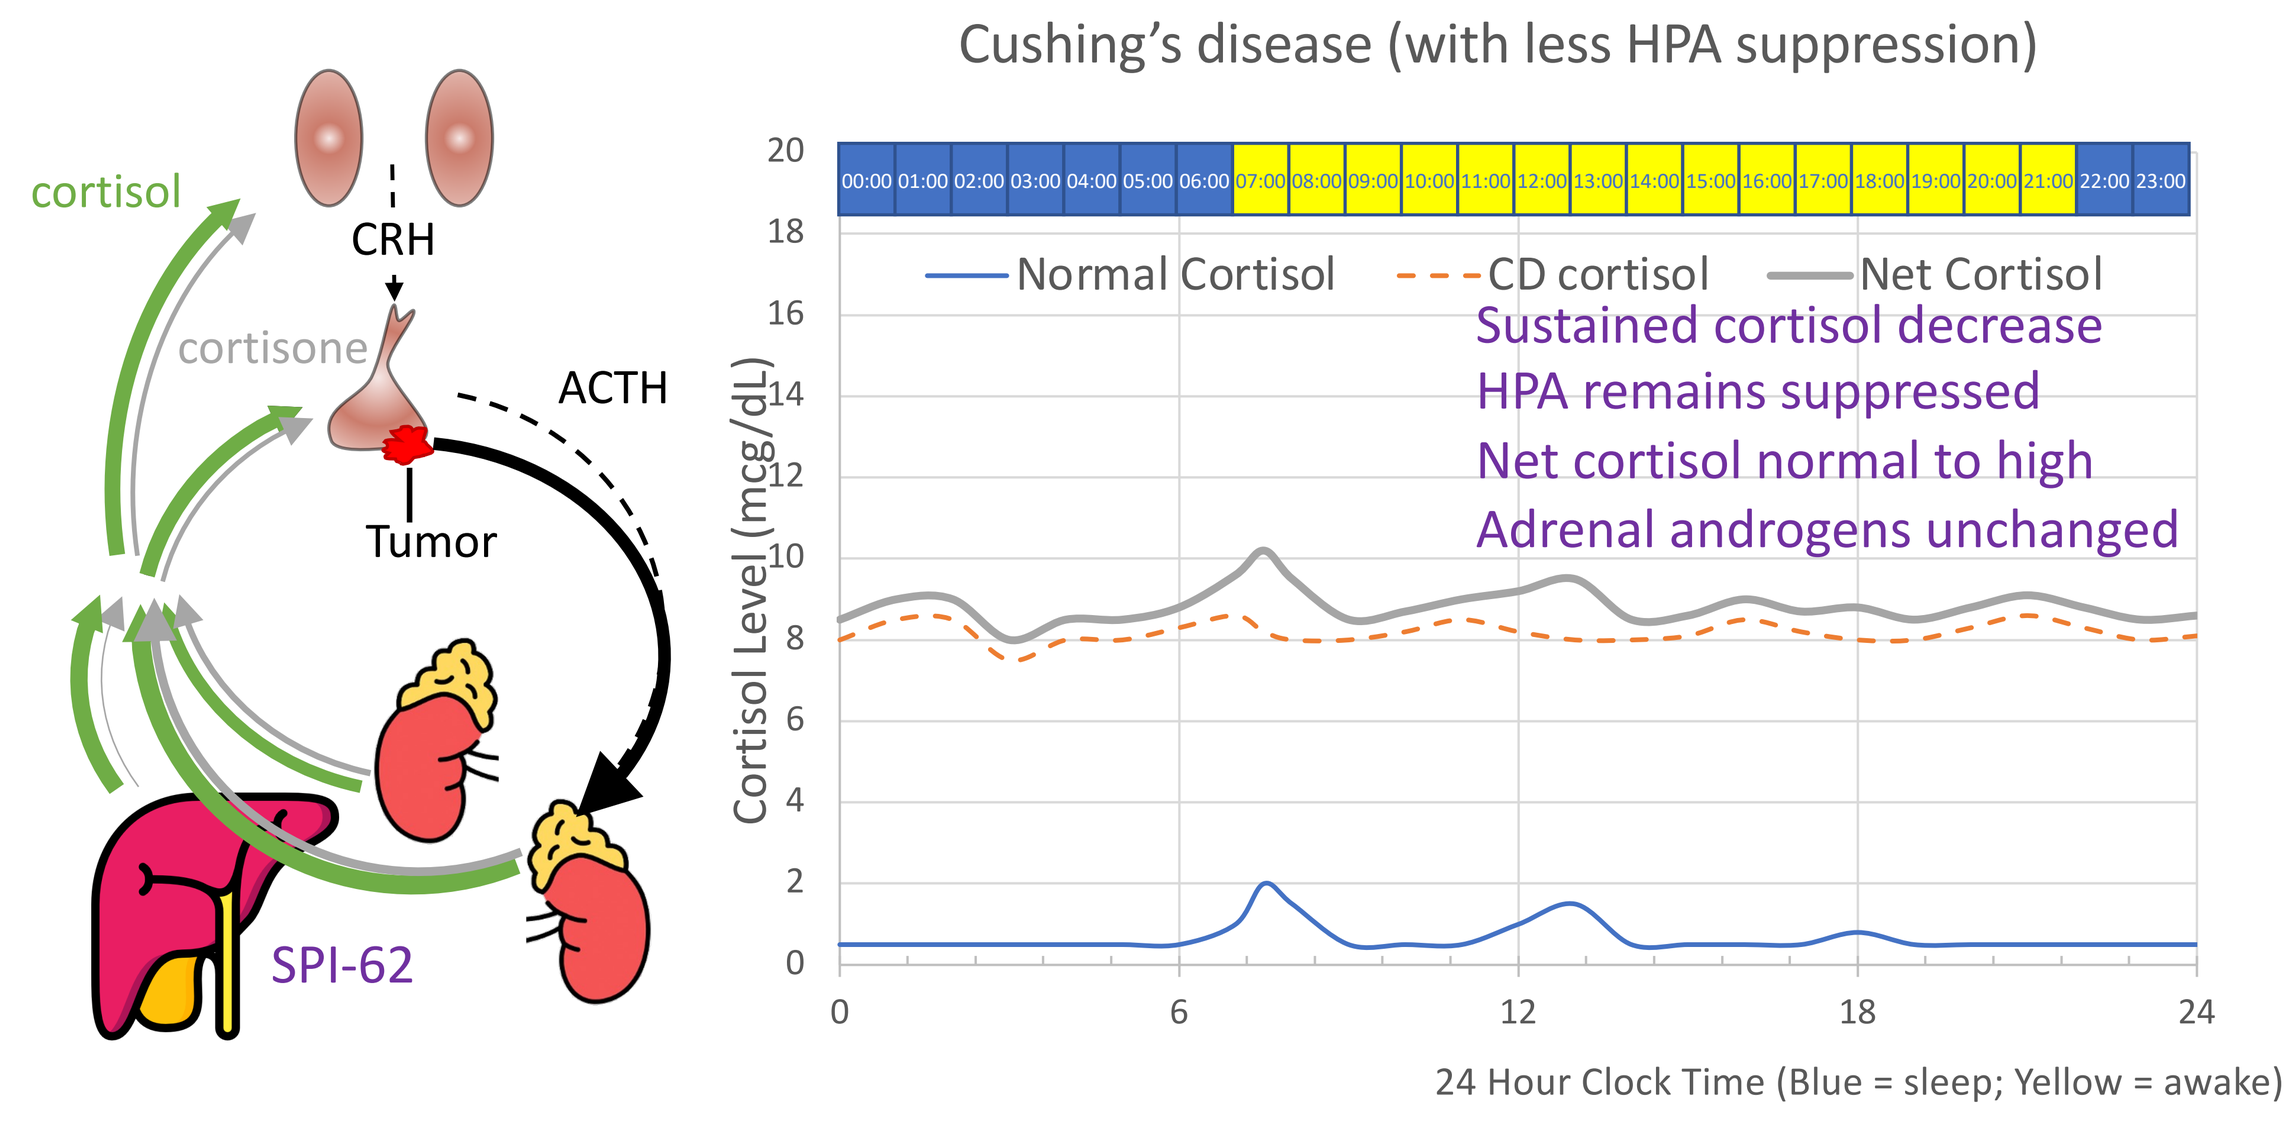
cortisol
CRH
cortisone
ACTH
Tumor
SPI-62
Cushing's disease (with less HPA suppression)
Normal Cortisol
CD cortisol
Net Cortisol
Sustained cortisol decrease
HPA remains suppressed
Net cortisol normal to high
Adrenal androgens unchanged
Cortisol Level (mcg/dL)
24 Hour Clock Time (Blue = sleep; Yellow = awake)

Adapted from Lacroix 2015 Lancet

SPI-62 effects on cortisol levels:

- In healthy adults, SPI-62 inhibition of HSD-1 reduces hepatocellular cortisol by 40-50% and (transiently) circulating cortisol by at least 30%. But within hours, HPA increases ACTH to restore circulating cortisol homeostasis.
- Whether HPA compensation will occur in patients with ACS is unknown. The degree of HPA suppression may be variable. SPI-62 might normalize cortisol levels and thereby reduce or eliminate cortisol morbidity.
- In patients with Cushing's syndrome, HPA compensation would not be expected due to non-tumorous corticotroph atrophy (unless driven by ectopic CRH secretion) or non-tumorous adrenal atrophy (in adrenal Cushing's syndrome or severe ACS). The magnitude of cortisol decrease might be larger than observed transiently in healthy adults. Both that and the degree to which SPI-62 might decrease cortisol morbidity remain to be determined.
- For patients administered SPI-62, cortisol stimulated or generated by the tumor should provide a sufficient buffer to preclude AI. However, large cortisol decreases on initiation might provoke glucocorticoid withdrawal syndrome (GWS).

Recognizing GWS:

- GWS symptoms are non-specific and may include:
 - Malaise, fatigue, lethargy, muscle weakness, diffuse myalgias or arthralgias, and fever
 - Nausea, abdominal pain, vomiting, anorexia, weight loss and postural hypotension (dizziness)
 - Dry flaking skin, psoriasis, connective tissue diseases, inflammatory bowel diseases
- More specific to GWS are:
 - Flu-like symptoms, irritability, mood swings, depression, anxiety, panic attacks, psychosis
- GWS is unlikely to be associated with:
 - Signs of AI such as infections, hyponatremia, hypoglycemia, and impaired consciousness

Recommendations for patients on HSD-1 inhibitors:

- Assess HPA function and the urinary HSD-1 ratio; in women, monitor for hyperandrogenic signs/symptoms (though biochemically should remain normal).
- Educate patients on symptoms and risks of GWS and AI during stress.
- Do not use prednisone or cortisone acetate as rescue medications; those are prodrugs requiring HSD-1 for activation.